A Multiomic, Single-Cell Measurable Residual Disease (scMRD) Assay For Phasing DNA Mutations and Surface Immunophenotypes

Poster #: AML-481

Charlie Murphy PhD*, Kathryn Thompson BS*, Lubna Nousheen MSc, Indira Krishnan PhD, Ben Geller BS, Aaron Llanso BS, Todd Druley MD, PhD, Daniel Mendoza PhD, Adam Sciambi PhD
Mission Bio. 400 E Jamie Ct, Suite 101, South San Francisco, CA 94080
* Contributed equally

Abstract

The small population of cancerous cells that remain following treatment, known as measurable residual disease (MRD), is the major cause of relapse in acute myeloid leukemia (AML). Usually, these refractory cells have gained additional resistance mutations or changed their surface immunophenotypes in ways that preclude detection and phasing by current gold standard flow cytometry or bulk next-generation sequencing assays. For this reason, a multiomic single-cell MRD (scMRD) assay could offer a more comprehensive indicator of relapse and the potential for faster response. Here, we present a new scMRD assay with a 0.01% limit of detection that provides single-cell clonal architecture and immunophenotyping to not only identify residual leukemia cells, but also identify putative DNA or protein targets for salvage therapy. The assay enables rare-cell detection on a standard Mission Bio Tapestri® run by adding (i) an upfront bead-based protocol to enrich for blast cells, (ii) a DNA and protein panel specifically designed for AML MRD diagnosis and treatment, and (iii) a new, automated analysis pipeline to evaluate single-cell multiomics output. By utilizing Mission Bio's technology for sequencing single cells, this pipeline can identify and correlate co-occurring de novo variants, thereby reducing false positive rates over bulk assays that do not correlate variants. It furthermore can create phylogenetic trees of the detected MRD cells and present their surface protein signature and arm-level copy number. In addition, the multiplexing of up to three patient samples combined in one run via germline identification further reduces per sample costs and increases throughput. To demonstrate these features on 0.01% MRD, samples were constructed by titrating cell lines or diseased cells into healthy bone marrow cells before processing them with the scMRD assay. We detected 0.01% and 0.1% spike-ins (CD34+ or CD117+) in 22 of 22 samples tested, with an average enrichment of 22.5x and 17x, respectively. We further applied the scMRD assay to banked bone marrow aspirate samples from 3 AML patients and achieved an average enrichment of 8.2x. The scMRD assay resolved the clonal architecture identifying multiple leukemic clones with co-occurring mutations. The assay readily distinguished pre-leukemic from leukemic clones thereby increasing the specificity of MRD results. The integration of genotype and immunophenotypic further enhanced MRD detection by identifying genotype-specific protein expression patterns. By combining high sensitivity with multiomics, this assay offers a potential scalable solution for comprehensive MRD detection that guides therapeutic decision-making.

Methods

Fig 1: scMRD assay workflow.

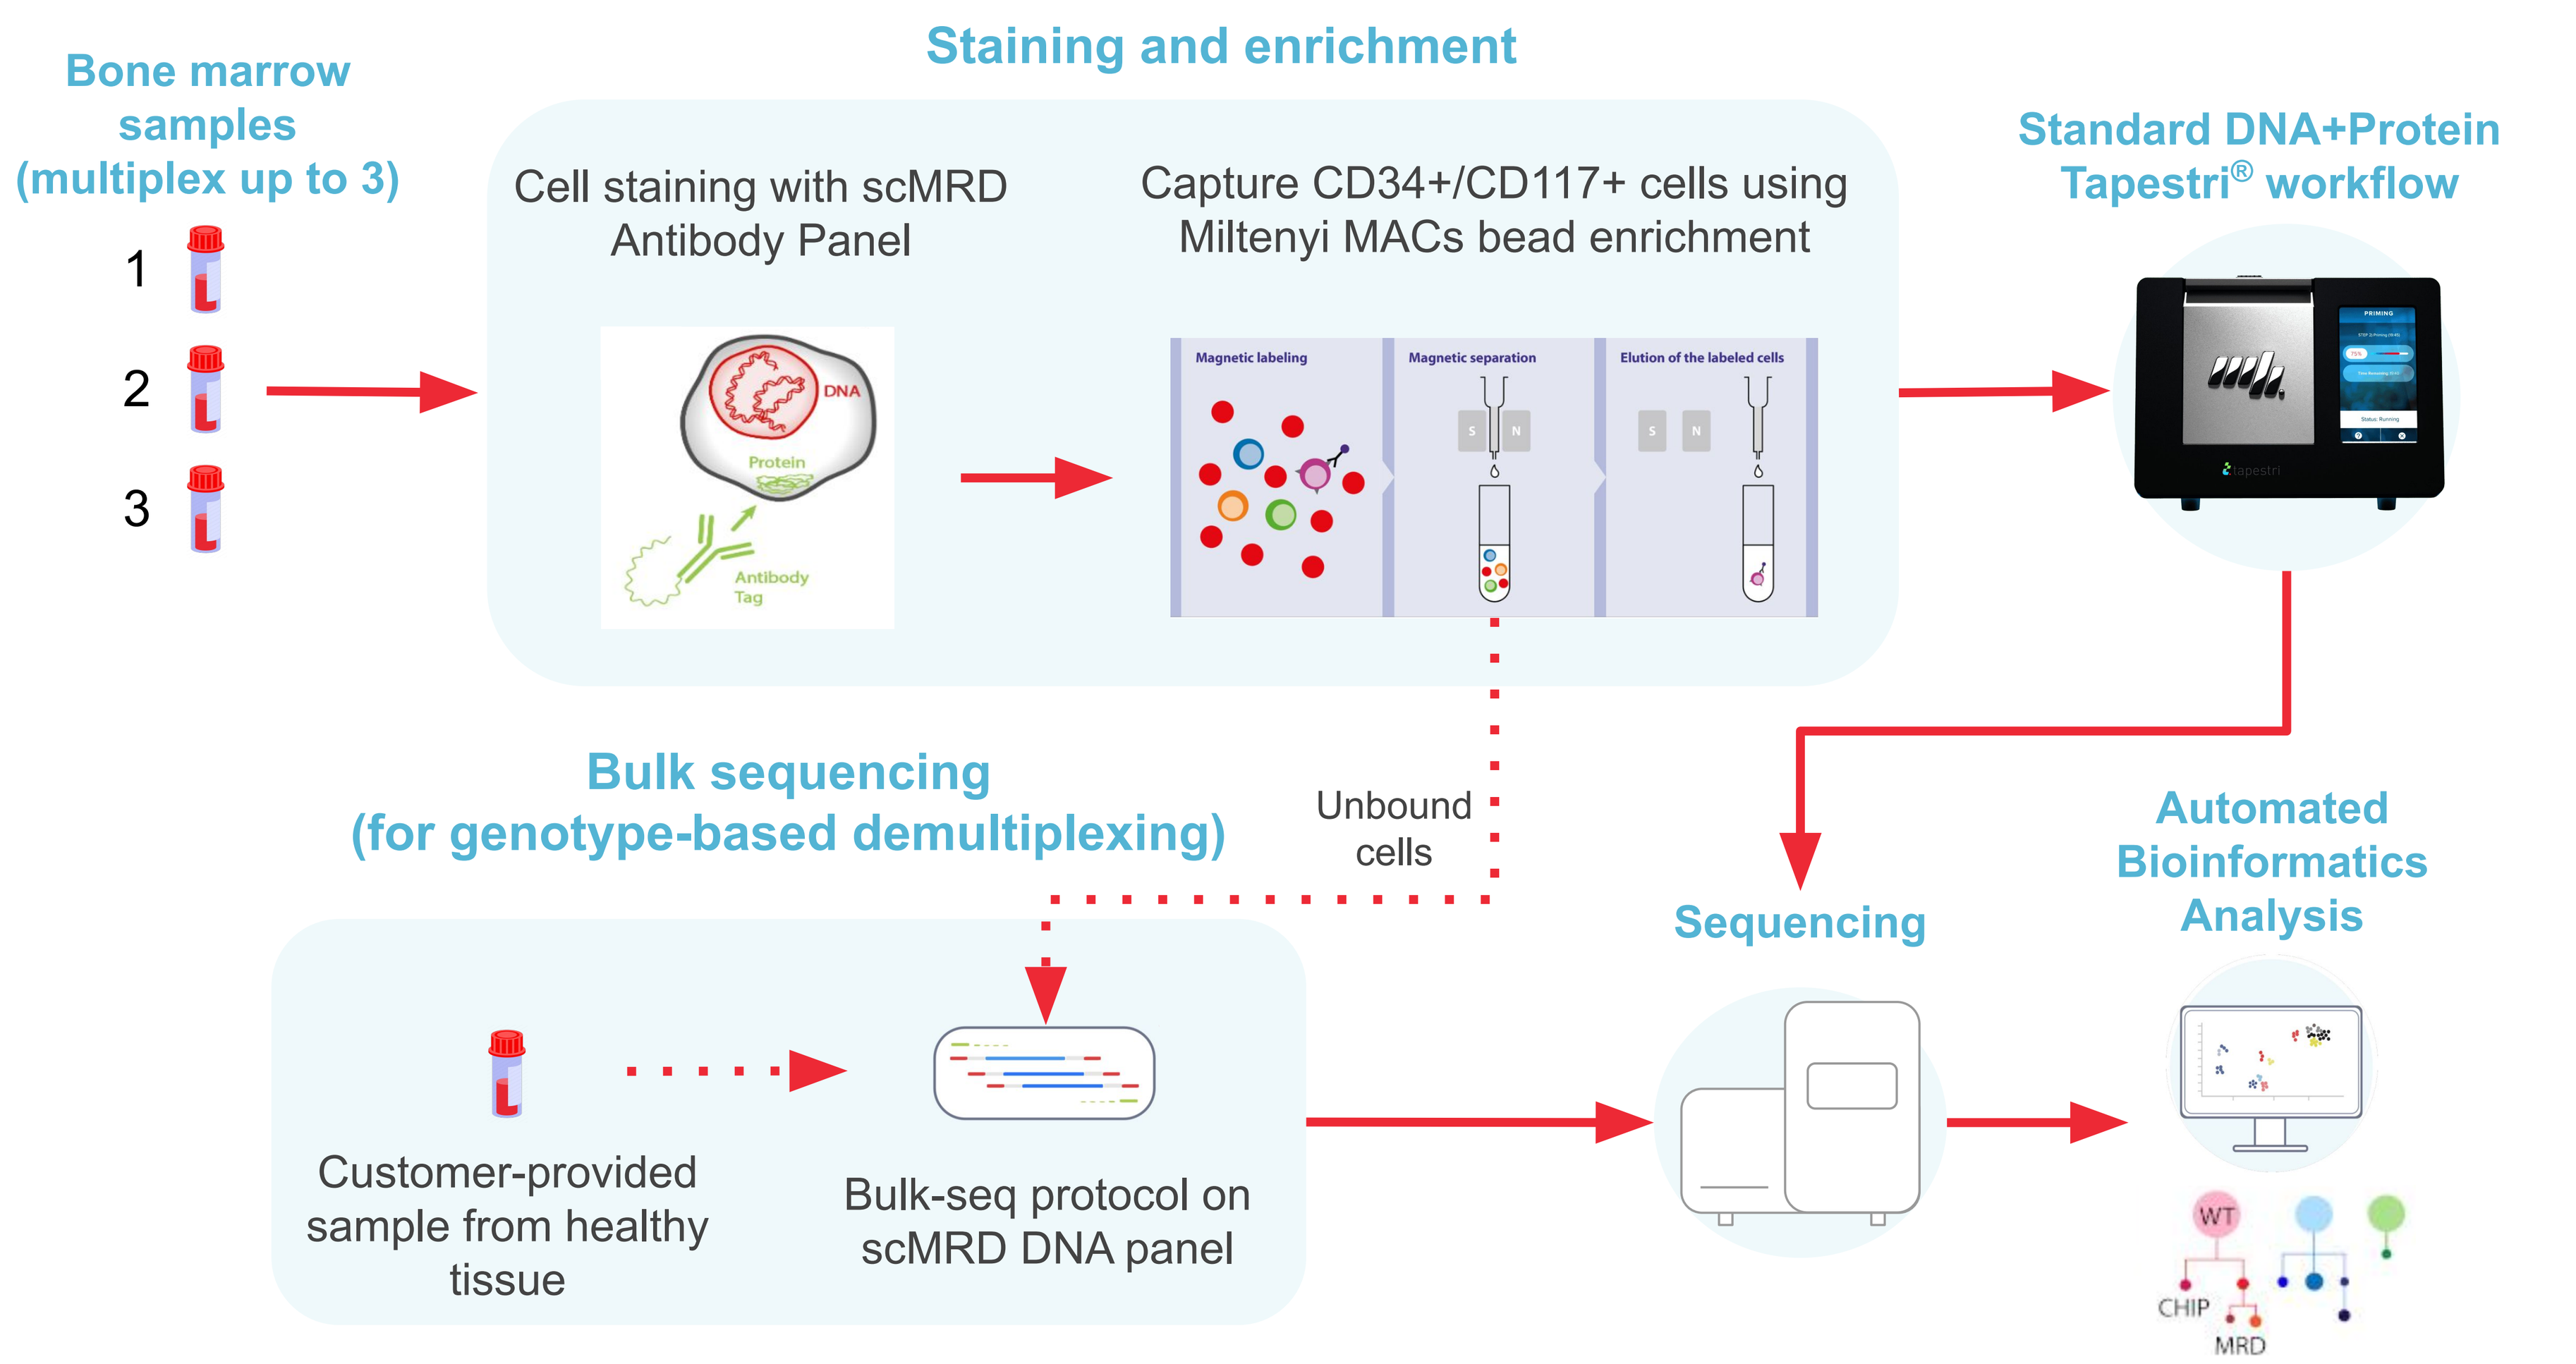
Bone marrow samples (multiplex up to 3)
1
2
3
Staining and enrichment
Cell staining with scMRD Antibody Panel
DNA
Protein
Antibody Tag
Capture CD34+/CD117+ cells using Miltenyi MACs bead enrichment
Magnetic labeling
Magnetic separation
Elution of the labeled cells
Standard DNA+Protein Tapestri® workflow
Unbound cells
Bulk sequencing (for genotype-based demultiplexing)
Customer-provided sample from healthy tissue
Bulk-seq protocol on scMRD DNA panel
Sequencing
Automated Bioinformatics Analysis
WT
CHIP
MRD

Tapestri® scMRD AML assay. The scMRD assay leverages the MACs bead enrichment protocol to enrich for CD34+ and/or CD117+ cells. Post enrichment, the samples are then processed using the standard Tapestri® DNA+Protein workflow. The assay allows multiplexing of up to 3 patient samples for a single assay run. The DNA panel (Figure 2 A) was designed to cover AML hotspot mutations and comply with European LeukemiaNet [1, 2] and International Consensus Classification [3] recommendations for AML MRD. The protein AOC panel was designed to include AML MRD disease specific biomarkers for immuno-phenotypic characterization of patient samples (Figure 2 C).
Assay performance. The assay performance was measured by using two sets of control cell lines that are CD34+ (KG1) or CD117+ (HMC-1.2), which have known variants orthogonally validated by bulk sequencing. These cell lines were spiked-in to healthy bone marrow cells as either 0.01% and 0.1%. These samples were then multiplexed up to three samples per run and prepared using the scMRD workflow. Similarly, performance was further assessed using clinical AML samples from diagnosis stage and spiked-in to healthy bone marrow cells from a different donor. Immunophenotype, phylogeny, and expected spike-in % were all confirmed with DNA and DNA+Protein Tapestri® runs without enrichment or dilution.
Bioinformatics. The assay includes a novel and automated workflow that takes as input FASTQs and outputs per-patient results. Demultiplexing uses each patient's known germline SNP genotypes. Rare variants are identified using novel algorithms that leverage the single-cell nature of co-occurring variants and per-variant background error rates based on control samples to reduce false positives.

Results

Fig 2: **(A)** Genes covered by the scMRD DNA panel. **(B)** The population frequency (gnomAD) of the 41 SNPs included in the scMRD DNA panel to capture the genetic diversity needed for demultiplexing. **(C)** The protein AOCs covered by the scMRD protein panel.

(A) DNA panel

ASXL1	FLT3	MYC	SF3B1
BCOR	GATA1	MYH11	SMC1A
BRAF	GATA2	NF1	SRSF2
CALR	IDH1	NPM1	STAG2
CBFB	IDH2	NRAS	TET2
CBL	IL6R*	PHF6	TP53
CHEK2	IP6K1*	PPM1D	TRPC4*
CSF1R	JAK2	PTPN11	U2AF1
CYP4F3*	KIT	RAD21	UBA1*
DNMT3A	KMT2A	RUNX1	WT1
ETV6	KRAS	SETBP1	ZEB2*
EZH2	MEIS2*	SF3A1*	ZRSR2

** For germline SNPs and sample demultiplexing*

(B)

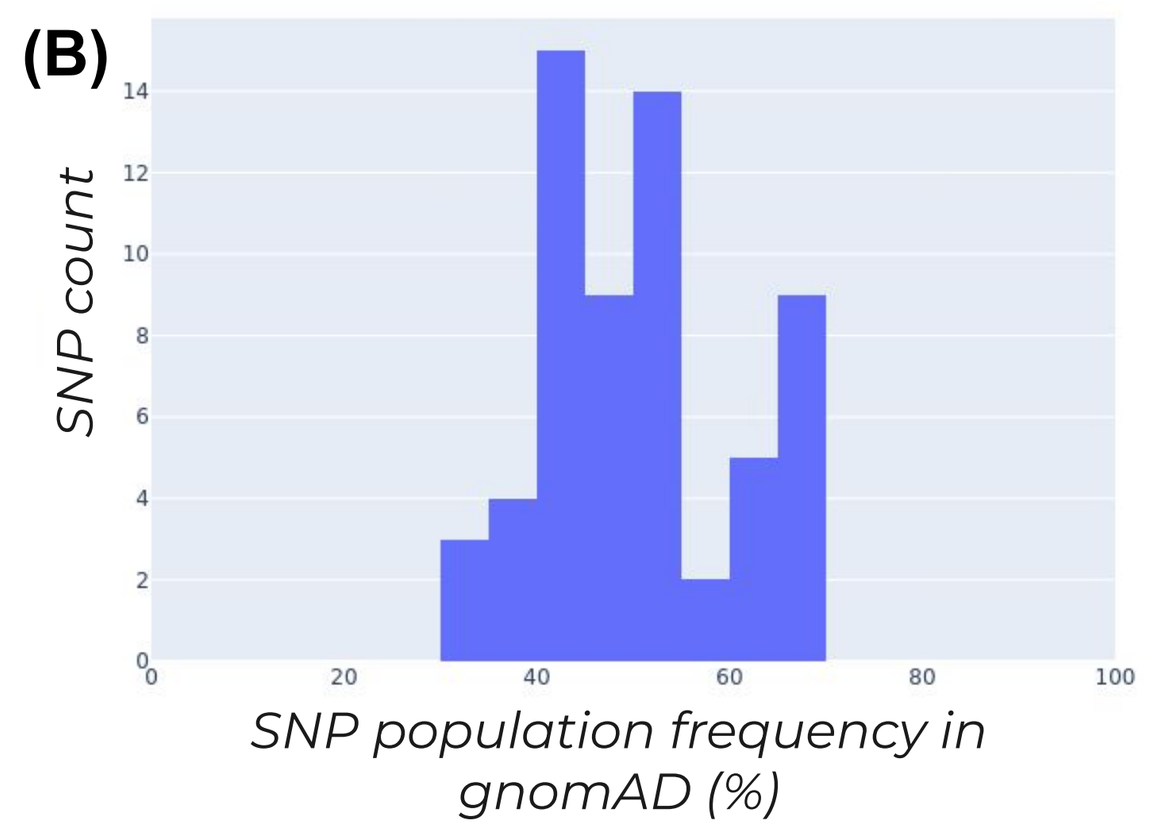
SNP count
SNP population frequency in gnomAD (%)

(C) Protein panel

Name	ID
CD2	0367 anti-human CD2
CD3	0034 anti-human CD3
CD7	0066 anti-human CD7
CD10	0062 anti-human CD10
CD11b	0161 anti-human CD11b
CD13	0364 anti-human CD13
CD14	0081 anti-human CD14
CD19	0050 anti-human CD19
CD22	0393 anti-human CD22
CD33	0052 anti-human CD33
CD34	0054 anti-human CD34
CD38	0389 anti-human CD38
CD45RA	0063 anti-human CD45RA
CD56	0047 anti-human CD56 (NCAM)
CD123	0064 anti-human CD123
HLA-DR	0159 anti-human HLA-DR
CD117	anti-human CD117 (A3C6E2)

Fig 3: Genotype and immunophenotype for each spike-in sample after CD34/CD117 enrichment (one per row). The genotypes shown are from a select number of high-coverage germline SNPs. For each sample, the spike-in clone with highest average expression of CD34 or CD117 is shown.

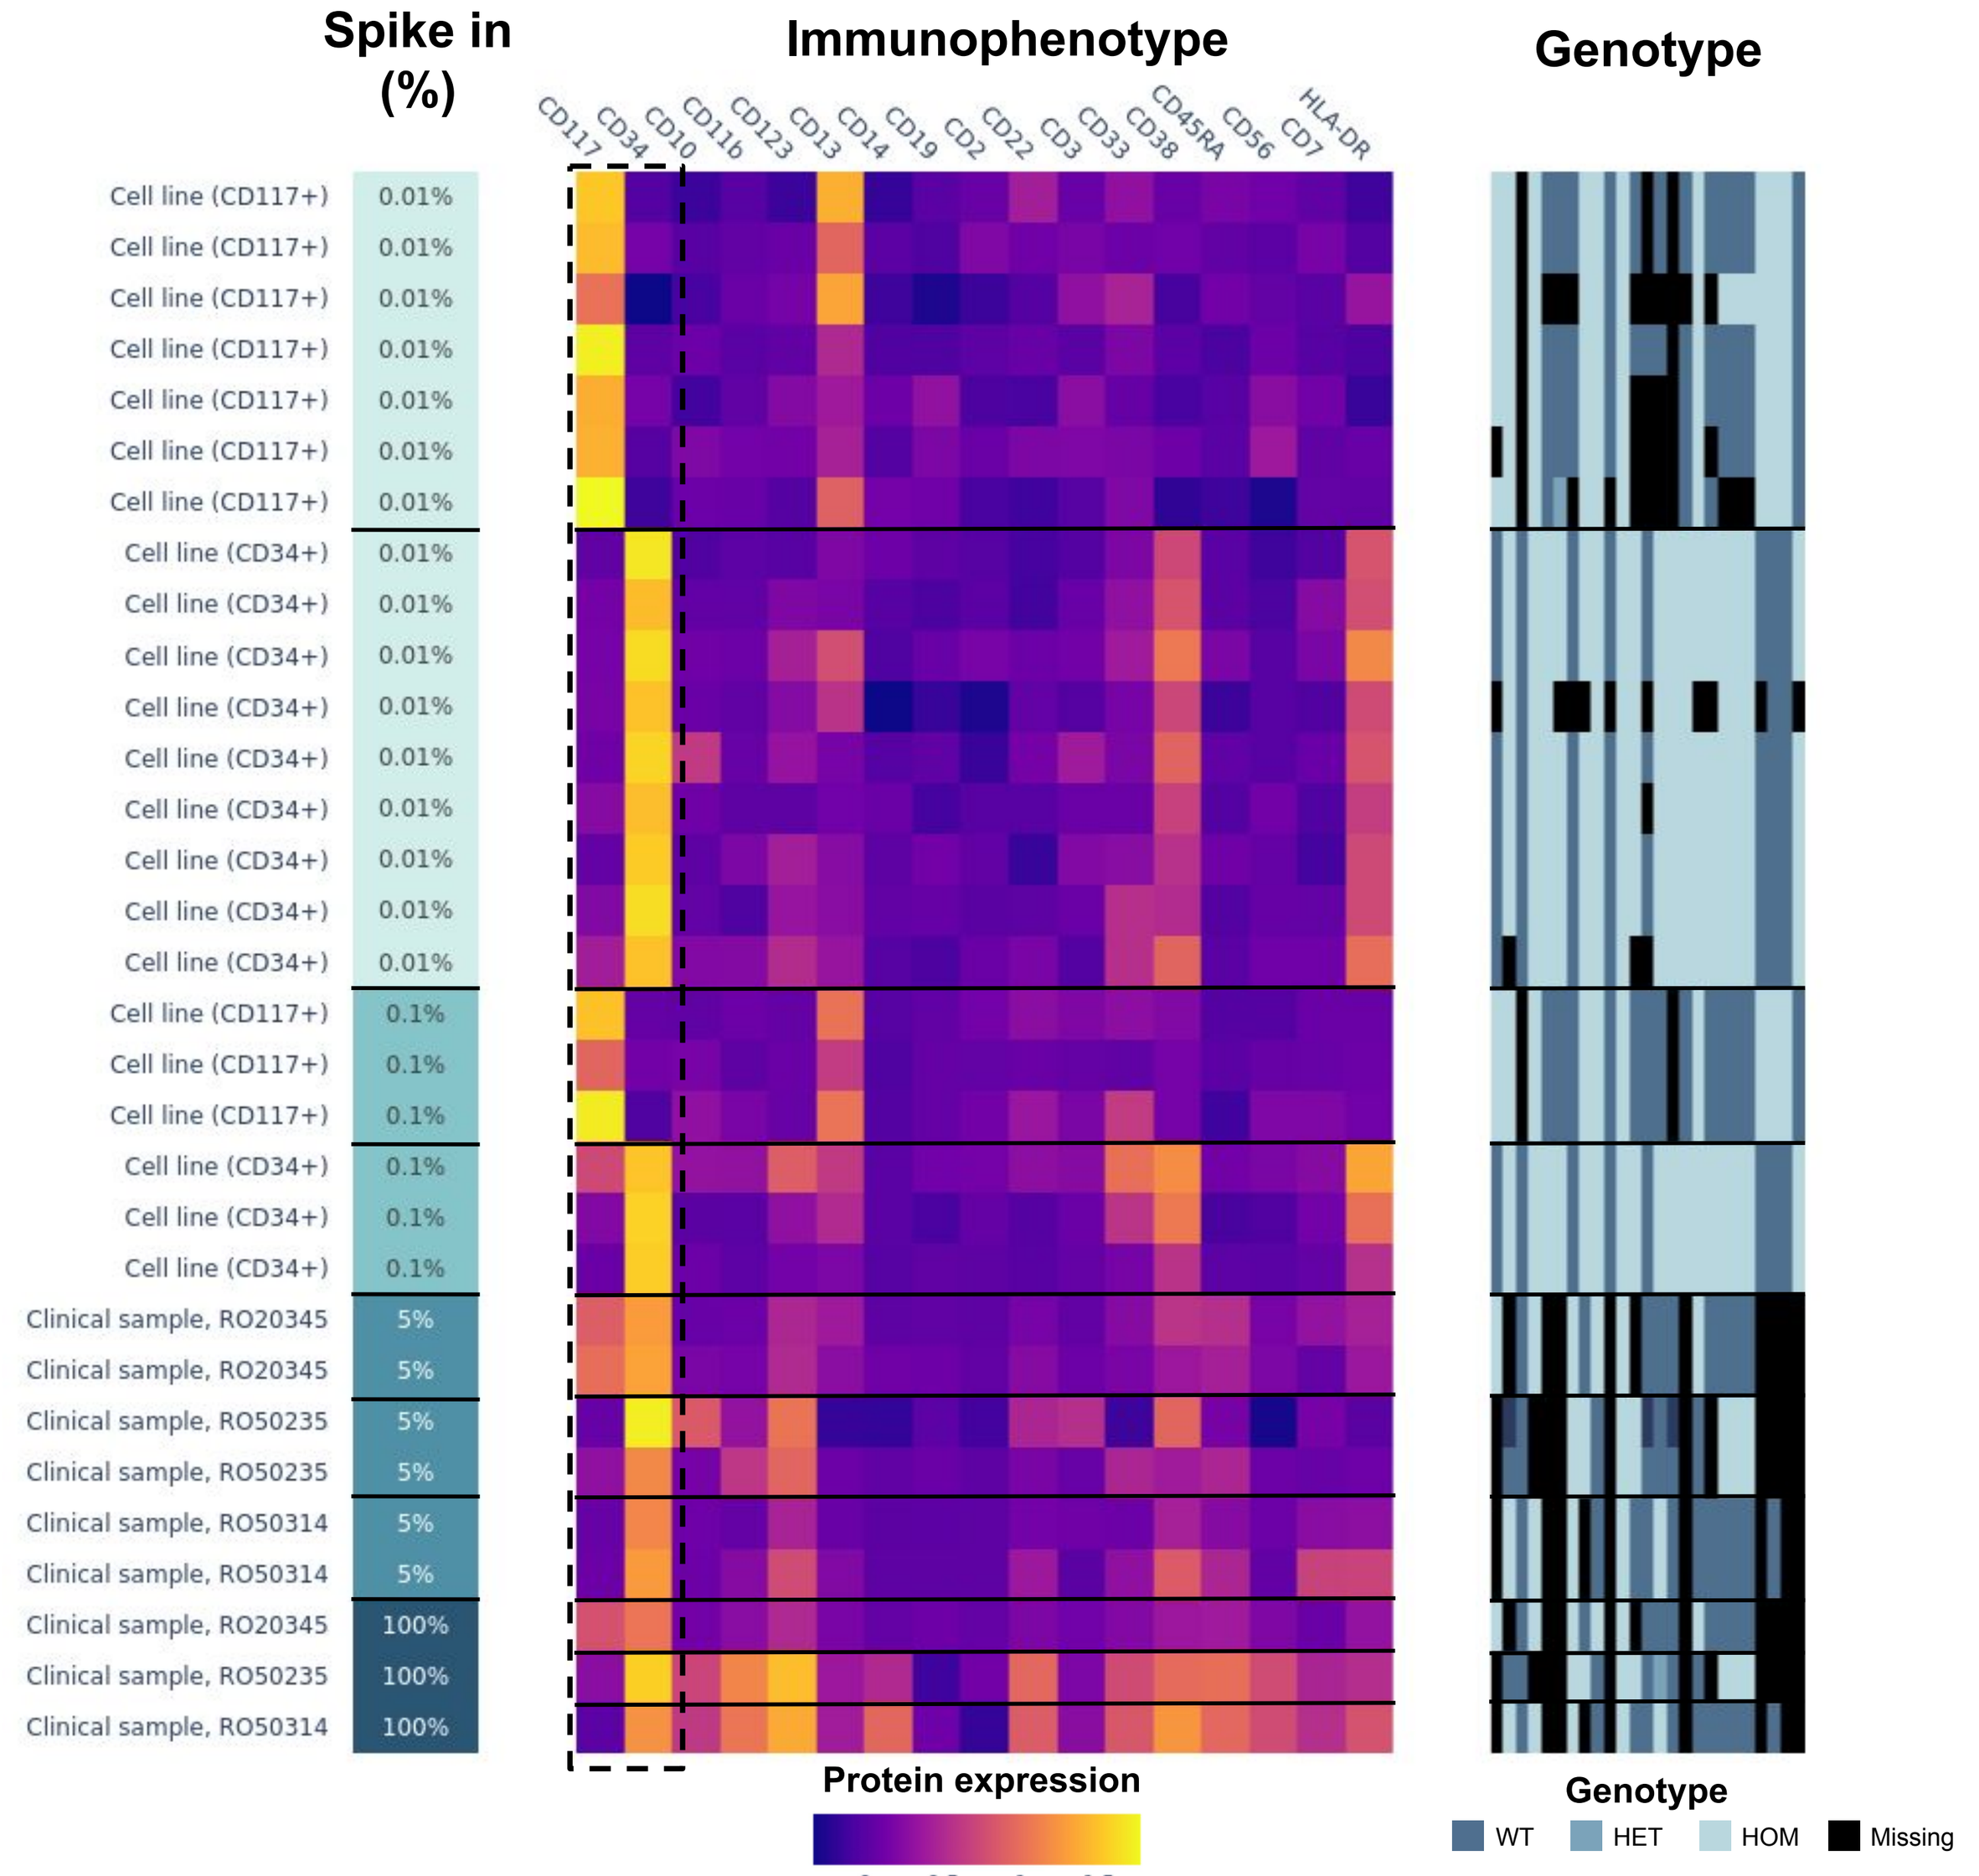
Spike in (%)
Immunophenotype
Genotype
CD117 CD34 CD10 CD11b CD123 CD13 CD14 CD19 CD2 CD22 CD3 CD33 CD38 CD45RA CD56 CD7 HLA-DR
Cell line (CD117+) 0.01%
Cell line (CD34+) 0.01%
Cell line (CD117+) 0.1%
Cell line (CD34+) 0.1%
Clinical sample, RO20345 5%
Clinical sample, RO50235 5%
Clinical sample, RO50314 5%
Clinical sample, RO20345 100%
Clinical sample, RO50235 100%
Clinical sample, RO50314 100%
Protein expression
0 0.5 1 1.5
Genotype
WT HET HOM Missing

Fig 4: **(A)** Example bioinformatics analysis of Tapestri® data from 1 clinical sample that contains 3 known somatic variants, where the corresponding somatic clones are CD34+/CD117+. **(B)** The corresponding flow cytometry data shows the abnormal blasts are CD34+/CD117+.

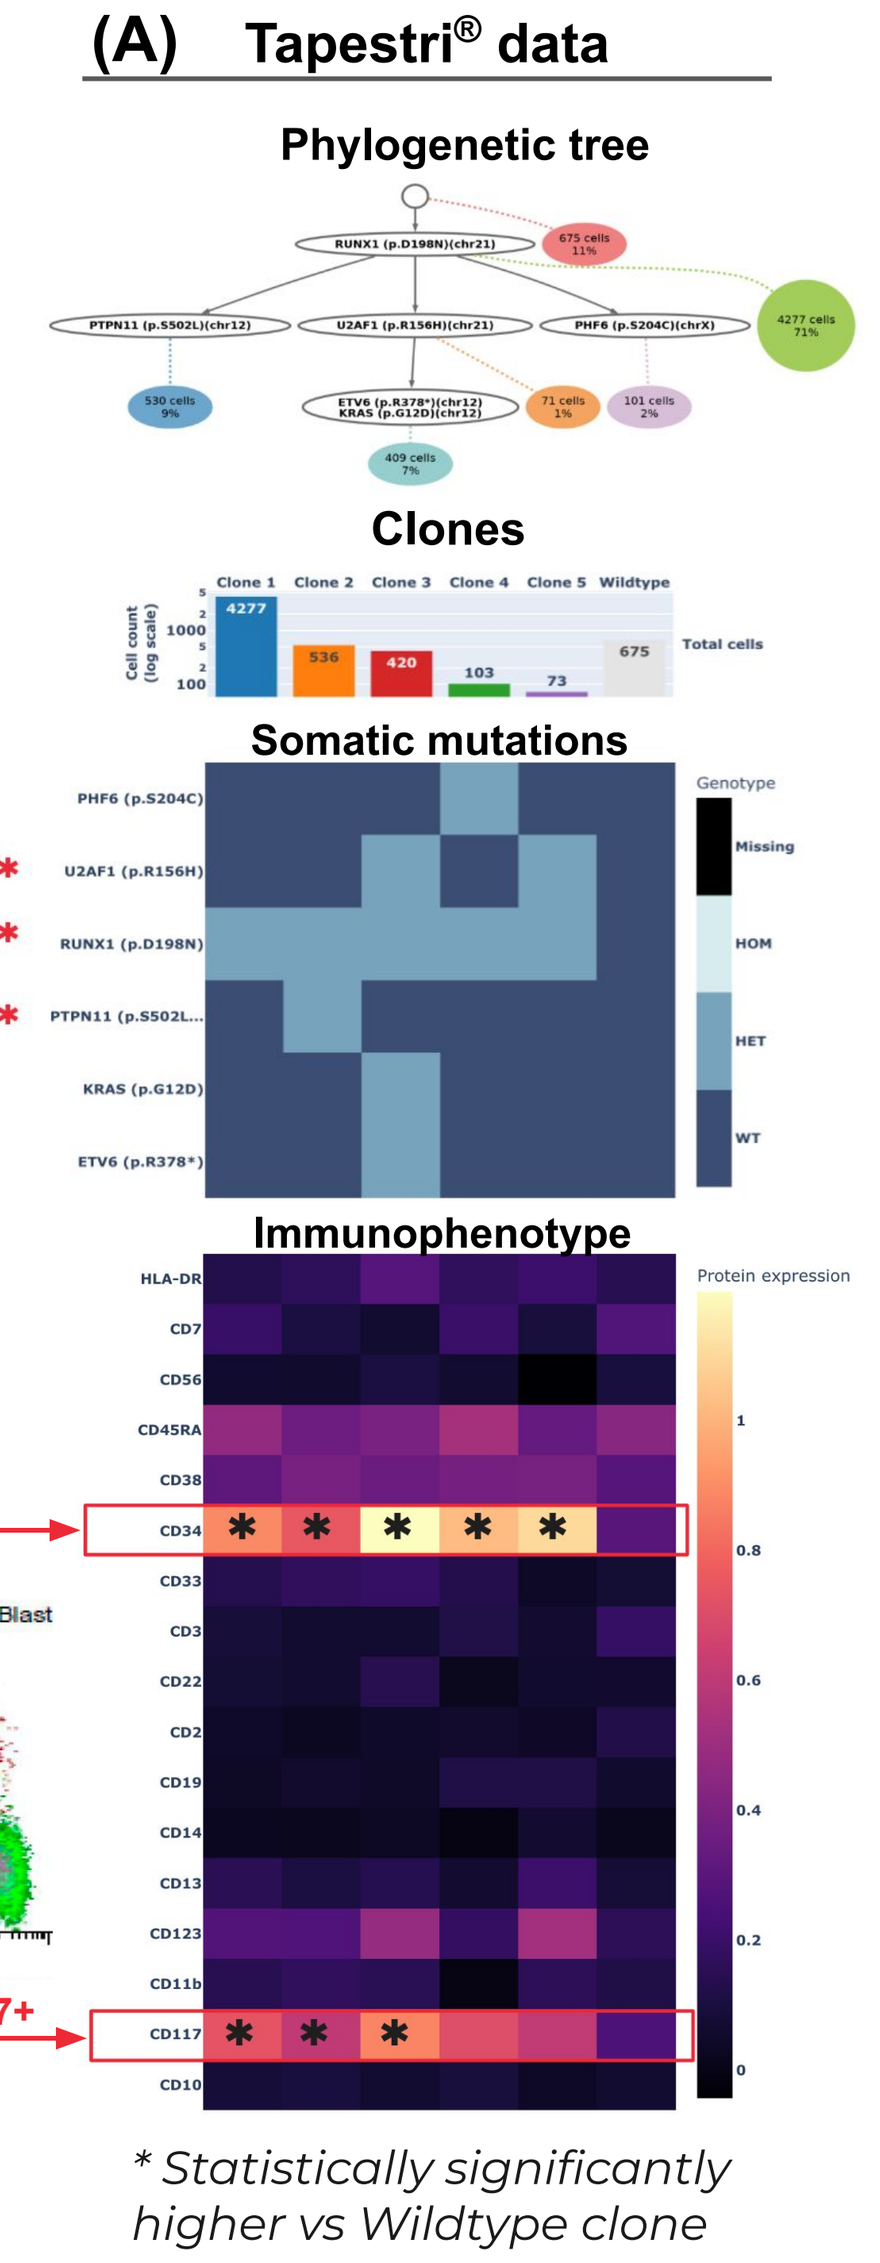
(A) Tapestri® data
Phylogenetic tree
Clones
Somatic mutations
Known somatic variants
Immunophenotype
Protein expression
* Statistically significantly higher vs Wildtype clone

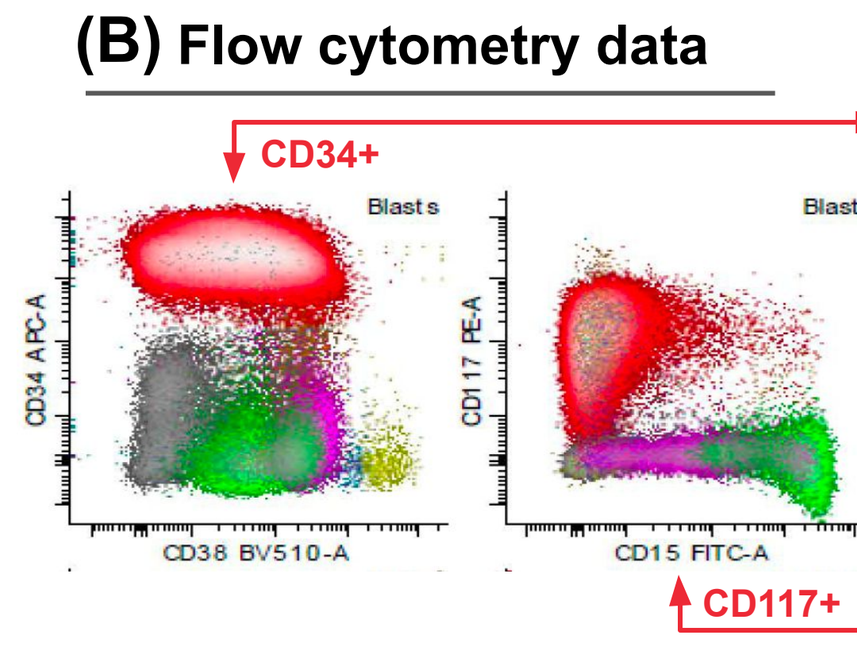
(B) Flow cytometry data
CD34+
CD117+
Blasts
Blast
CD34 APC-A
CD38 BV510-A
CD117 PE-A
CD15 FITC-A

Table 1: performance of the scMRD assay on samples containing artificial spike-ins from cell lines or clinical samples. The enrichment ratio is the ratio of the spike in % post enrichment divided by the spike-in % before enrichment.

Sample type	Immuno-phenotype	Spike-in %	Number of samples tested	Number of cells per multiplexed sample (average)	Number of spike-in cells (average)	Enrichment ratio (average)
Cell line (KG1)	CD34+	0.01%	9	4901	10.5	23.5
Cell line (KG1)	CD34+	0.1%	3	5130	113.3	21
Cell line (HMC-1.2)	CD117+	0.01%	7	5347	7.6	13.9
Cell line (HMC-1.2)	CD117+	0.1%	3	5121	104.3	20.1
Clinical sample	CD34+ &/ CD117+	5%	6	6314	2440.5	8.2

Table 2: performance of the scMRD bioinformatics pipeline on samples containing artificial spike-ins from cell lines or clinical samples. Spike in sensitivity is the percentage of samples where ≥1 known variant was detected.

Sample type	Immuno-phenotype	Spike-in %	Number of variants	Spike in sensitivity	Variant sensitivity (average)	Variant specificity (average)	False positive variants (average)
Cell line (KG1)	CD34+	0.01%	48	100%	79.5	99.9	0.22
Cell line (KG1)	CD34+	0.1%	48	100%	93.3	99.9	0.33
Cell line (HMC-1.2)	CD117+	0.01%	60	100%	67.1	99.9	0.29
Cell line (HMC-1.2)	CD117+	0.1%	60	100%	93.1	100	0
Clinical sample	CD34+ &/ CD117+	5%	11*	100%	95.8	99.9	1.5

* For clinical samples, we used orthogonally validated somatic variants. For cell lines, we used all known variants.

Fig 5: Enrichment protocol shows the expected change in cell type percentages in healthy bone marrow. Myeloid progenitors increase and T cells decrease. Each point is from a different Tapestri run. Sticky cells are those that express most proteins, and so are likely dead cells. Unknown are those that cannot be confidently assigned.

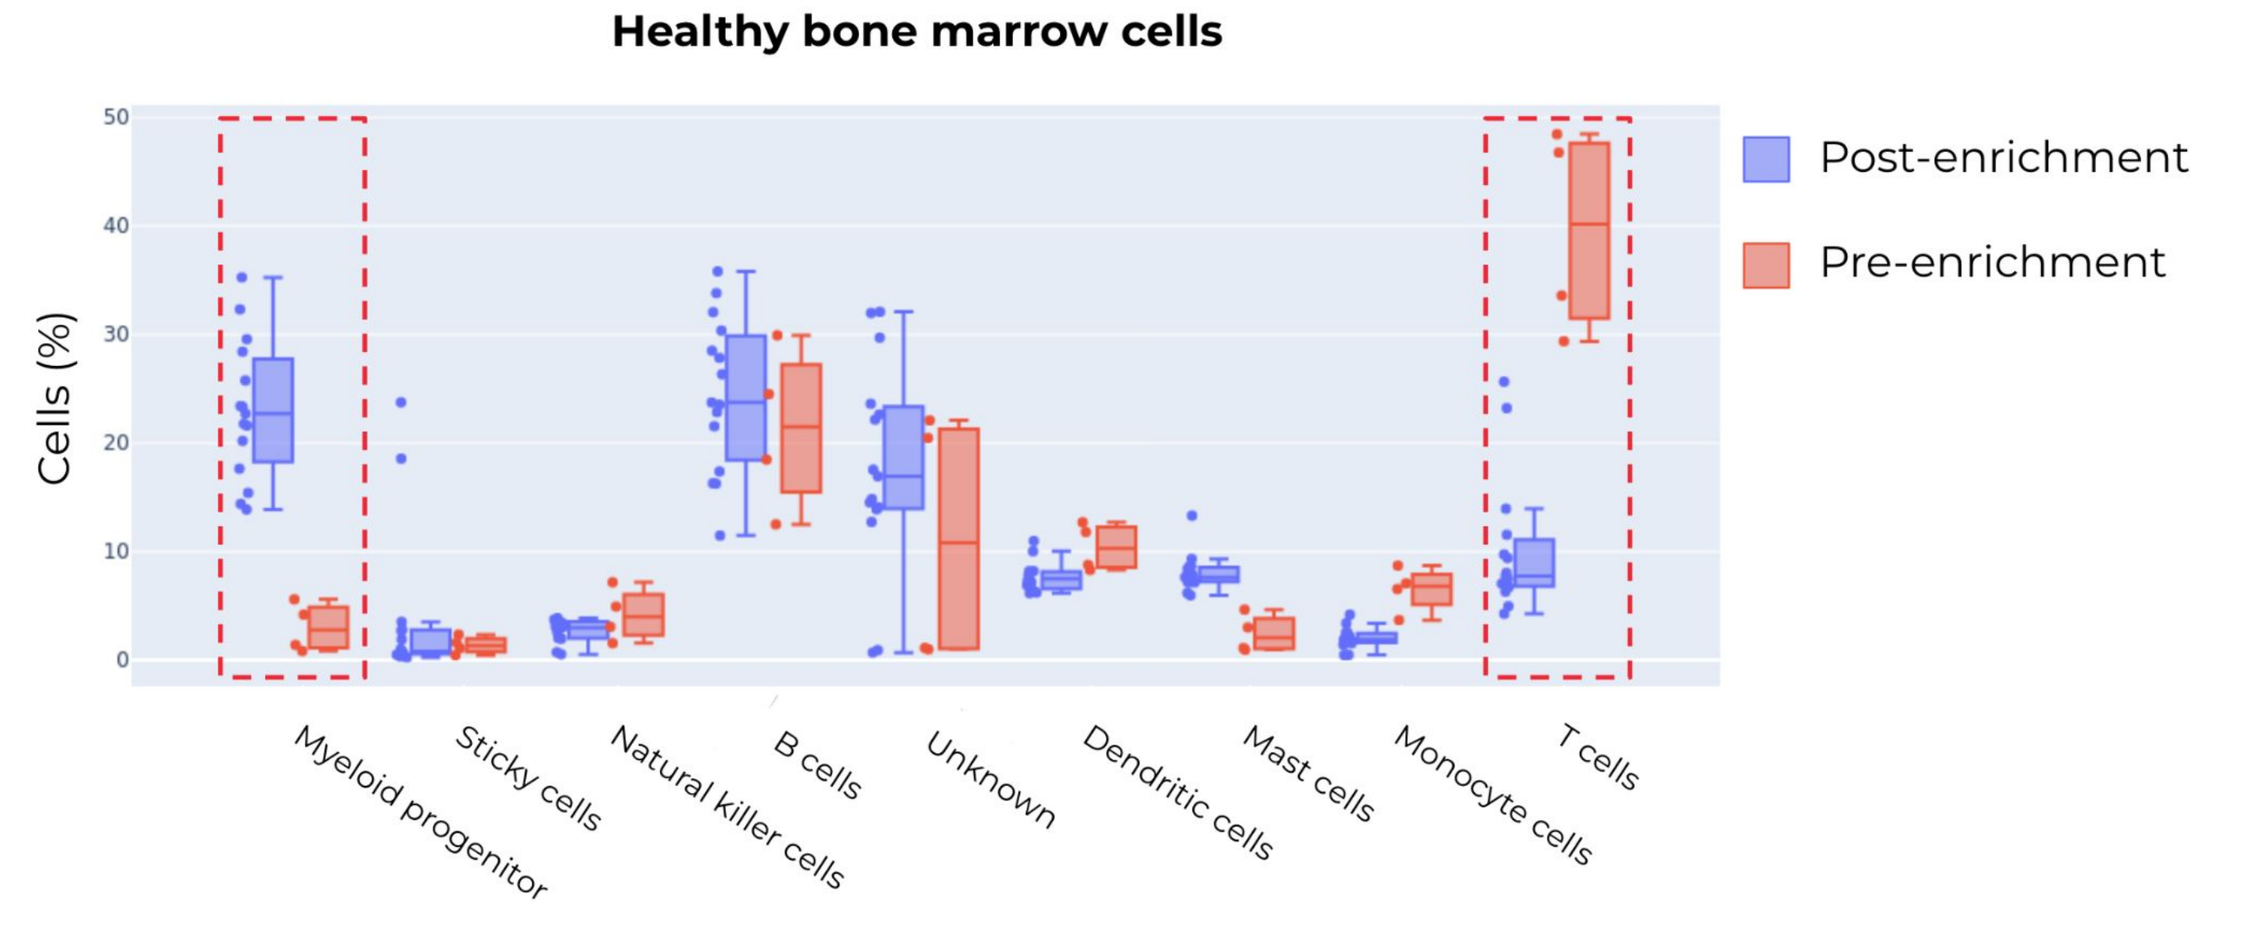
Healthy bone marrow cells
Cells (%)
Post-enrichment
Pre-enrichment
Myeloid progenitor
Sticky cells
Natural killer cells
B cells
Unknown
Dendritic cells
Mast cells
Monocyte cells
T cells

Conclusions

- Captured of >3 cells and detected ≥ 1 known variant in 100% of samples containing 0.01% or 0.1% spike-ins of CD34+ or CD117+ cell lines, with an average variant sensitivity of 83.4%.
- Detected 100% of clinical sample spike-ins at 5%, with an average variant sensitivity of 95%. Further testing on 0.01% and 0.1% spike-ins of clinical samples is pending.
- CD34+ and CD117+ cell line spike-ins were enriched on average 22.5x and 17x, respectively. Clinical samples were enriched on average 8.2x.
- Cell line spike-in phenotype matches expected CD34+ and CD117+ expression. Clinical sample phenotype aligns with flow data.
- Successfully demultiplexed up to 3 samples using bulk NGS.
- The new Tapestri® V3 chemistry yielded high cell capture rate (median of 19,637 cells per Tapestri® run).

References

1. Heuser, Michael, et al. "2021 Update on MRD in acute myeloid leukemia: a consensus document from the European LeukemiaNet MRD Working Party." Blood, The Journal of the American Society of Hematology 138.26 (2021): 2753-2767.
2. Döhner, Hartmut, et al. "Diagnosis and management of AML in adults: 2022 recommendations from an international expert panel on behalf of the ELN." Blood, The Journal of the American Society of Hematology 140.12 (2022): 1345-1377.
3. Arber, Daniel A., et al. "International Consensus Classification of Myeloid Neoplasms and Acute Leukemias: integrating morphologic, clinical, and genomic data." Blood, The Journal of the American Society of Hematology 140.11 (2022): 1200-1228.